Long-term Effect of Siponimod on MRI Outcomes in SPMS: Analyses from the EXPAND Study up to 5 Years

Douglas L. Arnold[1,2], Ludwig Kappos[3], Patrick Vermersch[4], Ralf Gold[5], Amit Bar-Or[6], Gavin Giovannoni[7], Bruce A.C. Cree[8], Daniela Piani Meier[9], Shannon Ritter[10], Goeril Karlsson[9], Frank Dahlke[9], Thomas Hach[9], Robert J. Fox[11]

Introduction

- Siponimod selectively modulates S1P (1,5) receptors[1,2] which are expressed on peripheral lymphocytes and within the central nervous system on the neurons and glial cells[3]
- Several preclinical studies have demonstrated that siponimod has direct effects in the CNS, eliciting beneficial effects on inflammation, neurodegeneration and remyelination[4–9]
- In the Phase 3 EXPAND core study, siponimod compared with placebo significantly reduced the risk of disability progression, showed benefits in cognitive processing speed, and MRI measures of focal inflammation and neurodegeneration (GM atrophy and MTR) in a broad population of patients with SPMS[10,11]

Objective

- Here, we use data from the core part+extension part of the EXPAND study to investigate the long-term effect of siponimod on MRI measures of inflammation and neurodegeneration including:
 - Lesion-based MRI parameters: change in T2LV and cumulative new/enlarging T2 lesions
 - Brain volume loss: total brain and grey matter (cGM and thalamic atrophy as measures of neurodegeneration)

Methods

- The core part was a variable treatment duration and event-driven study and of 1651 patients randomized in the core part (aged 18 to 60 years), 1224 entered the extension part
 - Patients receiving siponimod 2 mg/day continued to receive siponimod (continuous siponimod group)
 - Patients receiving placebo in core part were switched to receive siponimod 2 mg/day in the extension part (placebo-siponimod switch group).
- Given the event-driven design, time in the core part varied from 12 and up to 37 months for individual patients (median duration 21 months) before transitioning to the extension part (transition period between M12 and M36)
- **Between-group comparisons**: changes in MRI from core part baseline (M0) through M60 (core+extension parts) were compared between the continuous siponimod group and the placebo-siponimod switch group
- **Within-group comparisons**:* changes in MRI outcomes within each treatment group during the core part and during the extension part
 - To account for the variable exposure time, annualized measures of the MRI parameters were assessed for the first 12 months of core part and the first 12 months of extension part, as well as for the total duration of extension part (median 36 months) and total duration of core part (median 21 months):
 - Yearly average for T2LV change and new/enlarging T2 counts
 - Annualized rate of brain atrophy (ARBA) for rate of brain volume loss
- Treatment effects were determined using MMRM or Wilcoxon signed-rank test

*The percent change relative to start of EP was derived accounting for the change during the CP

cGM, cortical gray matter; CP, core part; CNS, central nervous system; EP, extension part; GM, gray matter; M, months; MMRM, mixed model of repeated measures; MRI, magnetic resonance imaging; MTR, magnetization transfer ratio; SPMS, secondary progressive multiple sclerosis; S1P, sphingosine 1-phosphate; T2LV, T2 lesion volume

1. Scalfari A, et al. J Neurol Neurosurg Psychiatry 2014, 85: 67-75; 2. Neuenschwander B, et al. Biometrics 2020, 76: 578-587; 3. Colombo E, et al. Front Immunol 2020, 11:635; 4. Choi JW and Chun J. Biochim Biophys Acta 2013, 1831: 20–32; 5. Gentile A, et al. J Neuroinflammation 2016, 13: 207; 6. Jaillard C, et al. J Neurosci 2005, 25: 1459–1469; 7. Mannioui A, et al. Mult Scler 2018, 24: 1421–1432; 8. O'Sullivan C, et al. J Neuroinflammation 2016, 13: 31; 9. Jackson SJ, et al. J Neuroinflammation 2011, 8: 76; 10. Kappos L, et al. Lancet 2018, 391: 1263–1273; 11. Benedict RHB, et al. Neurology 2021, 96: e376–e386

Arnold et al.

Results

Between-group comparison: Placebo-siponimod vs continuous siponimod from CP baseline by timepoint

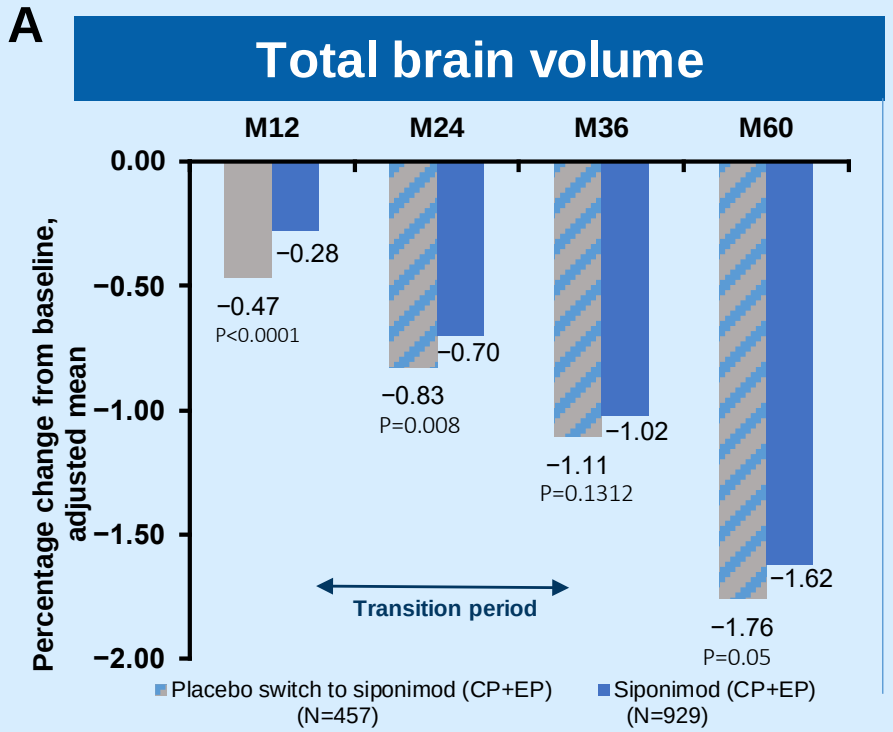
A
Total brain volume
M12
M24
M36
M60
0.00
−0.50
−1.00
−1.50
−2.00
Percentage change from baseline, adjusted mean
−0.28
−0.47
P<0.0001
−0.70
−0.83
P=0.008
−1.02
−1.11
P=0.1312
−1.62
−1.76
P=0.05
Transition period
Placebo switch to siponimod (CP+EP) (N=457)
Siponimod (CP+EP) (N=929)

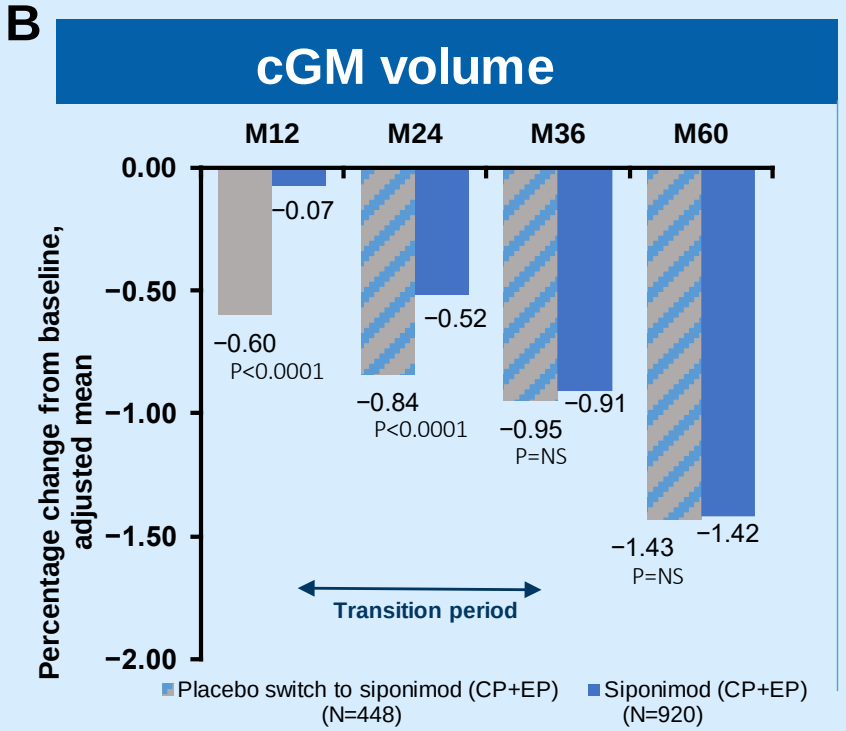
B
cGM volume
M12
M24
M36
M60
0.00
−0.50
−1.00
−1.50
−2.00
Percentage change from baseline, adjusted mean
−0.07
−0.60
P<0.0001
−0.52
−0.84
P<0.0001
−0.91
−0.95
P=NS
−1.42
−1.43
P=NS
Transition period
Placebo switch to siponimod (CP+EP) (N=448)
Siponimod (CP+EP) (N=920)

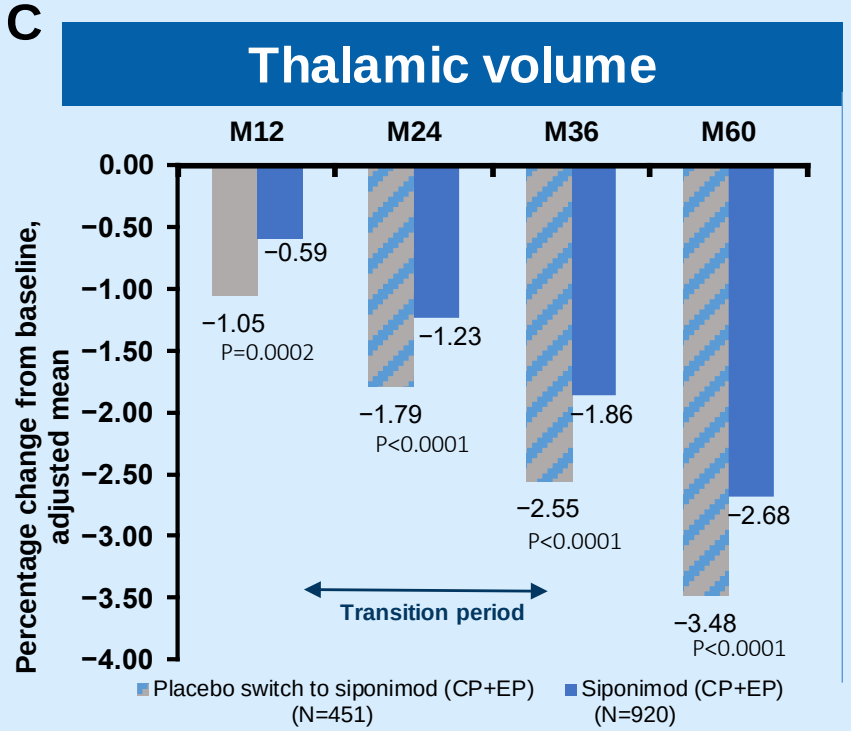
C
Thalamic volume
M12
M24
M36
M60
0.00
−0.50
−1.00
−1.50
−2.00
−2.50
−3.00
−3.50
−4.00
Percentage change from baseline, adjusted mean
−0.59
−1.05
P=0.0002
−1.23
−1.79
P<0.0001
−1.86
−2.55
P<0.0001
−2.68
−3.48
P<0.0001
Transition period
Placebo switch to siponimod (CP+EP) (N=451)
Siponimod (CP+EP) (N=920)

Placebo before switch to siponimod (M12)

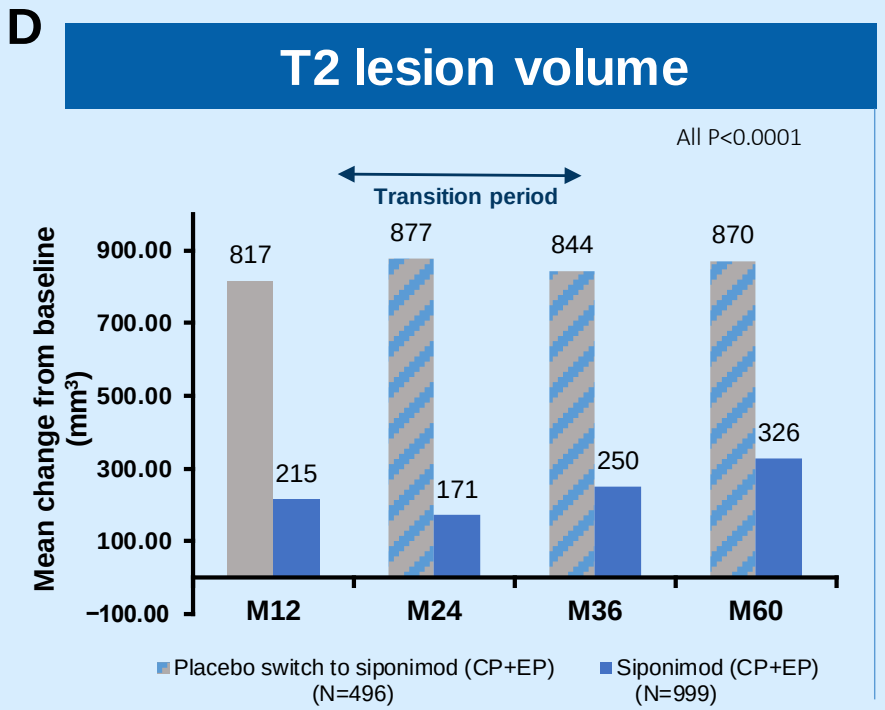
D
T2 lesion volume
All P<0.0001
Transition period
Mean change from baseline (mm³)
900.00
700.00
500.00
300.00
100.00
−100.00
817
215
877
171
844
250
870
326
M12
M24
M36
M60
Placebo switch to siponimod (CP+EP) (N=496)
Siponimod (CP+EP) (N=999)

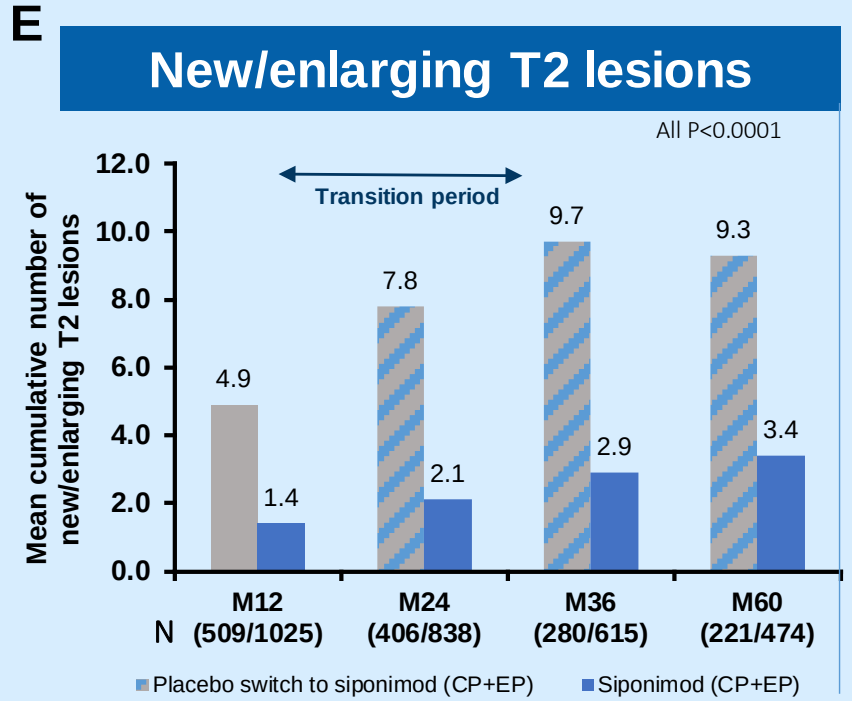
E
New/enlarging T2 lesions
All P<0.0001
Transition period
Mean cumulative number of new/enlarging T2 lesions
12.0
10.0
8.0
6.0
4.0
2.0
0.0
4.9
1.4
7.8
2.1
9.7
2.9
9.3
3.4
N
M12 (509/1025)
M24 (406/838)
M36 (280/615)
M60 (221/474)
Placebo switch to siponimod (CP+EP)
Siponimod (CP+EP)

- Persistent differences up to M60 favoring continuous siponimod vs the placebo switch group were observed on thalamic and total brain volume as well as T2LV and the number of new/enlarging T2 lesions
- For cGM, volume loss remained low in both treatment groups suggesting an immediate benefit upon switch

A–D: MMRM model — percent change from baseline adjusted for visit, treatment, age, number of Gd+ T1 lesions at baseline, T2 lesion volume (cm^3) at baseline, superimposed relapses at baseline, visit*treatment interaction (and baseline normalized brain tissue volume, where applicable);
E: p-values from Wilcoxon rank sum test for differences between treatment groups

cGM, cortical gray matter; CP, core part; EP, extension part; Gd, Gadolinium; M, months; MMRM, mixed model for repeated measures; MRI, magnetic resonance imaging, N, number of patients; NS, not significant; T2LV, T2 lesion volume

Arnold et al.

Results

Within-group comparison: Placebo-siponimod switch group

A Total brain volume

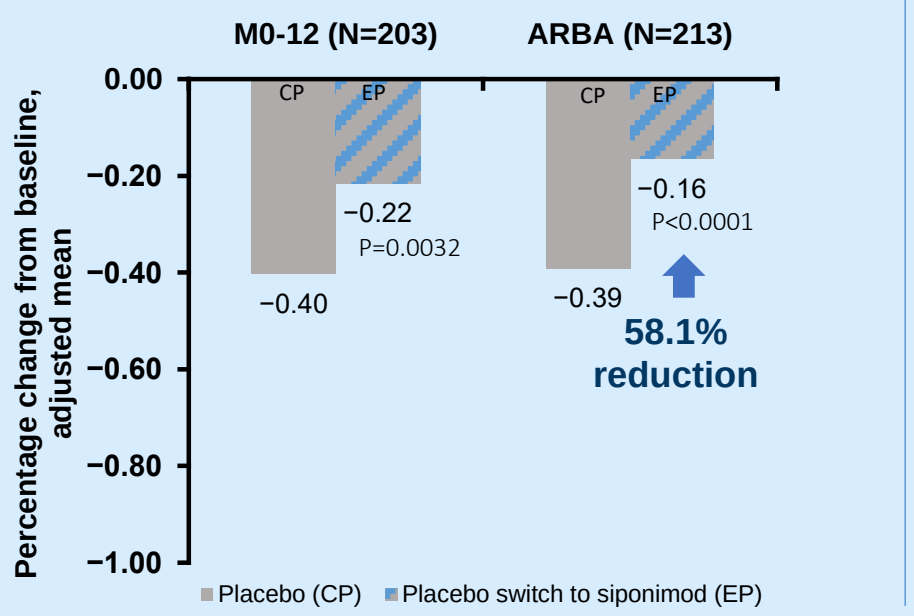
M0-12 (N=203)
ARBA (N=213)
Percentage change from baseline, adjusted mean
0.00
−0.20
−0.40
−0.60
−0.80
−1.00
CP
EP
−0.40
−0.22
P=0.0032
−0.39
−0.16
P<0.0001
58.1% reduction
Placebo (CP)
Placebo switch to siponimod (EP)

B cGM volume

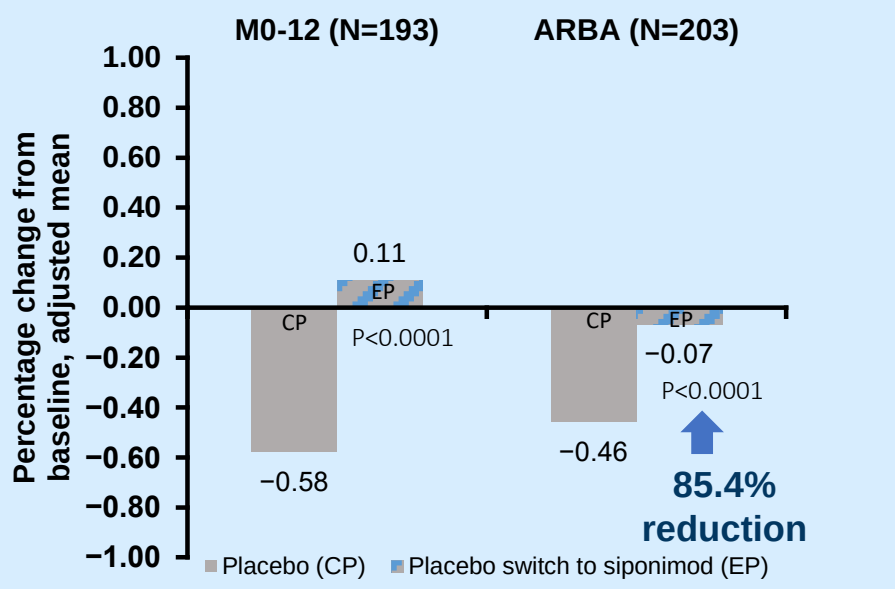
M0-12 (N=193)
ARBA (N=203)
Percentage change from baseline, adjusted mean
1.00
0.80
0.60
0.40
0.20
0.00
−0.20
−0.40
−0.60
−0.80
−1.00
CP
EP
0.11
P<0.0001
−0.58
−0.46
−0.07
P<0.0001
85.4% reduction
Placebo (CP)
Placebo switch to siponimod (EP)

C Thalamic volume

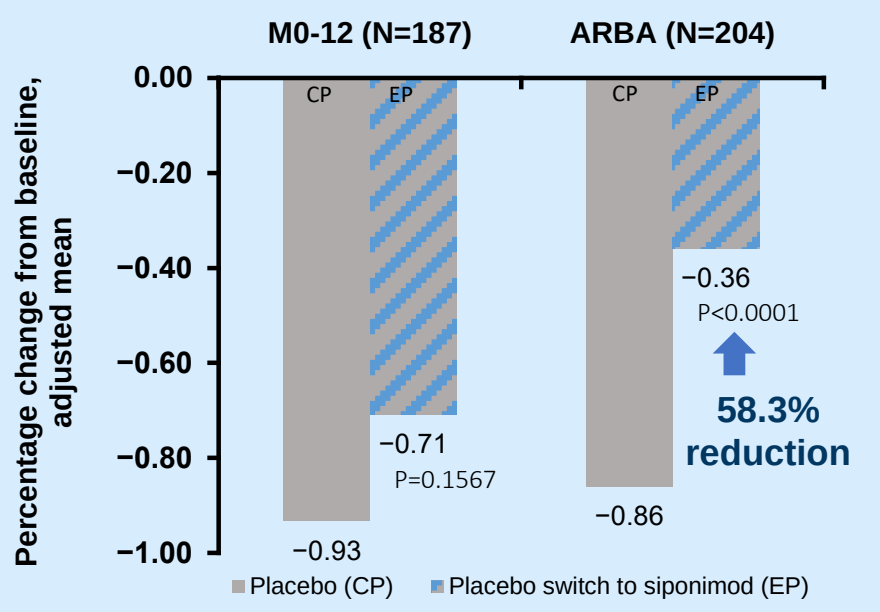
M0-12 (N=187)
ARBA (N=204)
Percentage change from baseline, adjusted mean
0.00
−0.20
−0.40
−0.60
−0.80
−1.00
CP
EP
−0.93
−0.71
P=0.1567
−0.86
−0.36
P<0.0001
58.3% reduction
Placebo (CP)
Placebo switch to siponimod (EP)

ARBA is derived from percent change to last visit during CP (median 21 months) and during EP (median 36 months)

D T2 lesion volume

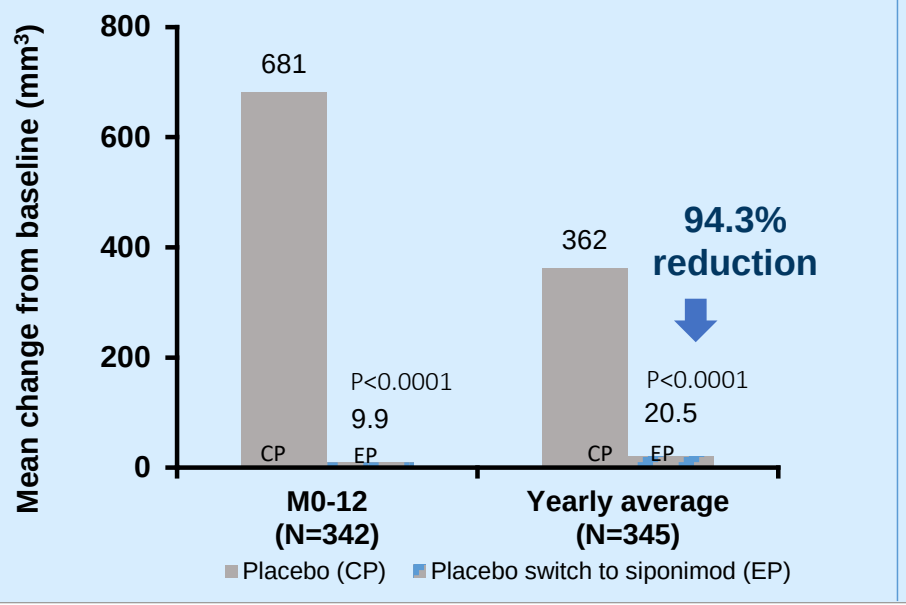
Mean change from baseline (mm³)
800
600
400
200
0
681
CP
EP
P<0.0001
9.9
362
P<0.0001
20.5
94.3% reduction
M0-12 (N=342)
Yearly average (N=345)
Placebo (CP)
Placebo switch to siponimod (EP)

E New/enlarging T2 lesions

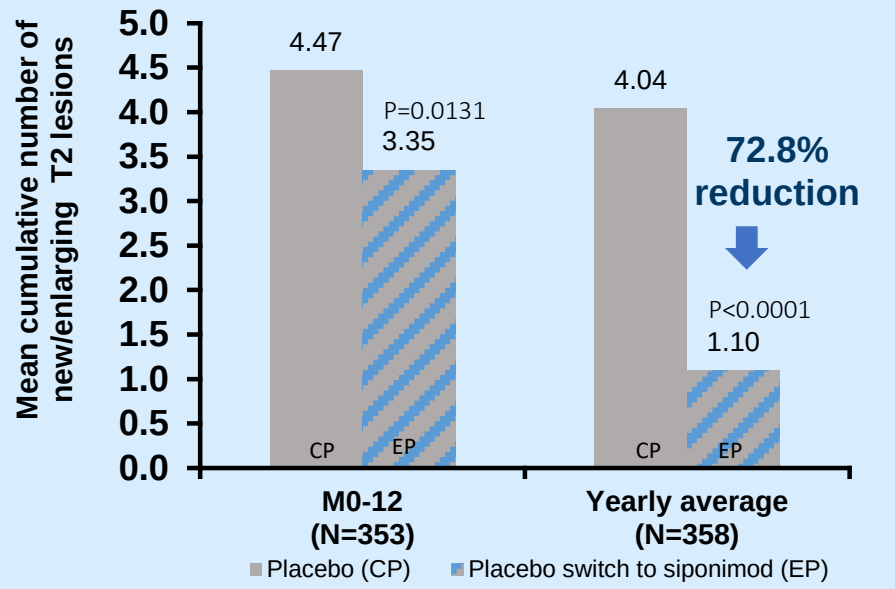
Mean cumulative number of new/enlarging T2 lesions
5.0
4.5
4.0
3.5
3.0
2.5
2.0
1.5
1.0
0.5
0.0
4.47
P=0.0131
3.35
4.04
72.8% reduction
P<0.0001
1.10
CP
EP
M0-12 (N=353)
Yearly average (N=358)
Placebo (CP)
Placebo switch to siponimod (EP)

- A significant improvement was observed on all MRI measures upon switching from placebo in the core part to siponimod in the extension part
- There was a complete suppression of cGM atrophy and no increase in T2 lesion volume within the first 12 months upon switch from placebo to siponimod (M0–12 of the extension part)

At each time point, only patients with a value both at the CP visit and the corresponding EP visit are included; the yearly average is derived by standardizing the change to the last visit to 360 days
p-values from Wilcoxon signed-rank test comparing percent changes between CP and EP within each group

ARBA, annualized rate of brain atrophy; cGM, cortical grey matter; CP, core part; EP, extension part; M, months; MRI, magnetic resonance imaging; N, number of patients; T2LV, T2 lesion volume

Arnold et al.

Results

Within-group comparison: Continuous siponimod group

A

Total brain volume

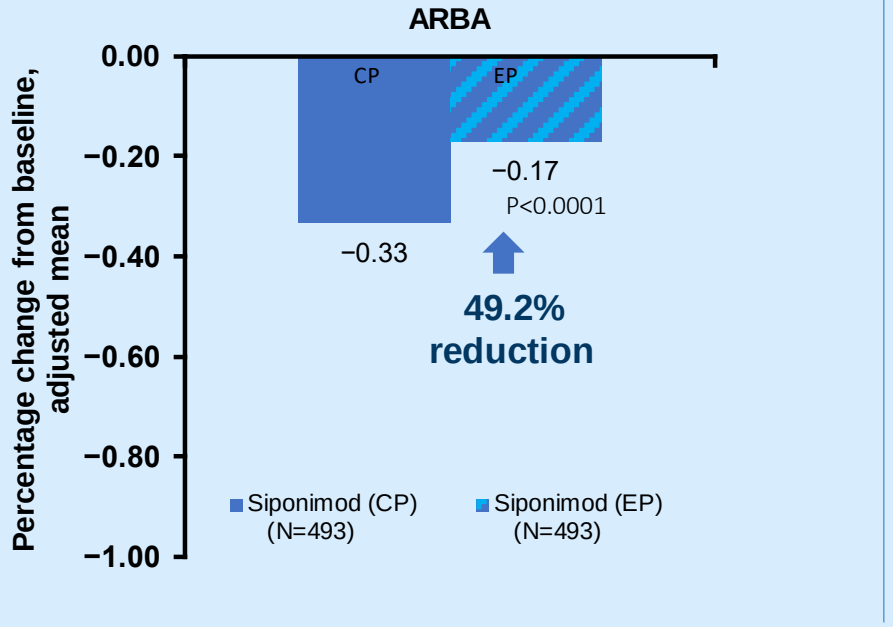
ARBA
Percentage change from baseline, adjusted mean
0.00
−0.20
−0.40
−0.60
−0.80
−1.00
CP
EP
−0.33
−0.17
P<0.0001
49.2% reduction
Siponimod (CP) (N=493)
Siponimod (EP) (N=493)

B

cGM volume

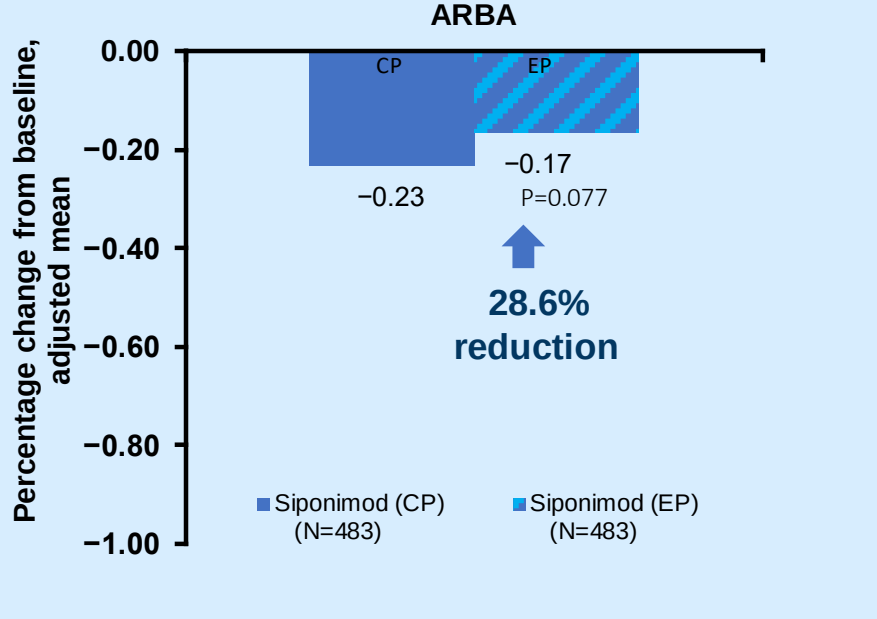
ARBA
Percentage change from baseline, adjusted mean
0.00
−0.20
−0.40
−0.60
−0.80
−1.00
CP
EP
−0.23
−0.17
P=0.077
28.6% reduction
Siponimod (CP) (N=483)
Siponimod (EP) (N=483)

C

Thalamic volume

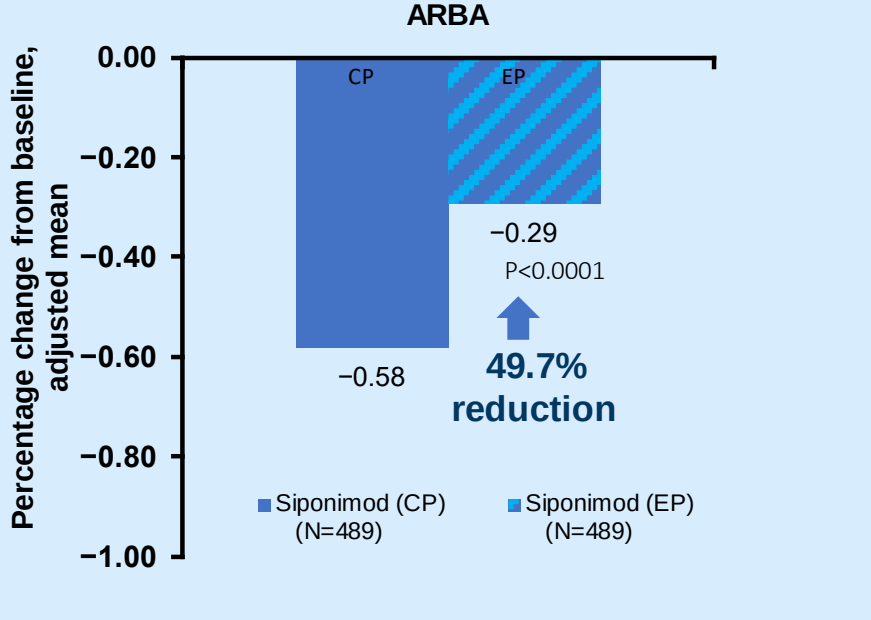
ARBA
Percentage change from baseline, adjusted mean
0.00
−0.20
−0.40
−0.60
−0.80
−1.00
CP
EP
−0.58
−0.29
P<0.0001
49.7% reduction
Siponimod (CP) (N=489)
Siponimod (EP) (N=489)

ARBA is derived from percent change to last visit during CP (median 21 months) and during EP (median 36 months)

D

T2 lesion volume

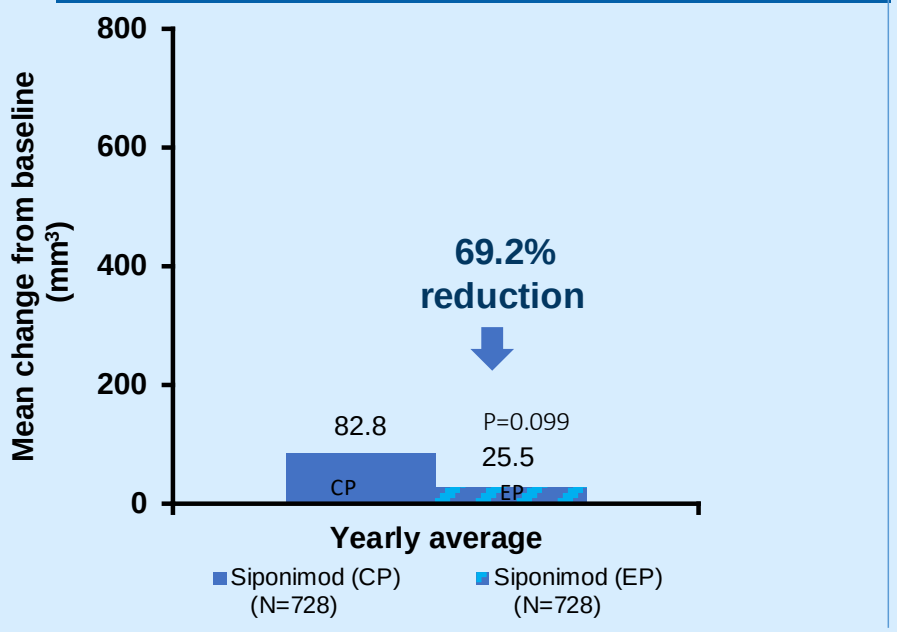
Mean change from baseline (mm³)
800
600
400
200
0
69.2% reduction
82.8
P=0.099
25.5
CP
EP
Yearly average
Siponimod (CP) (N=728)
Siponimod (EP) (N=728)

E

New/enlarging T2 lesions

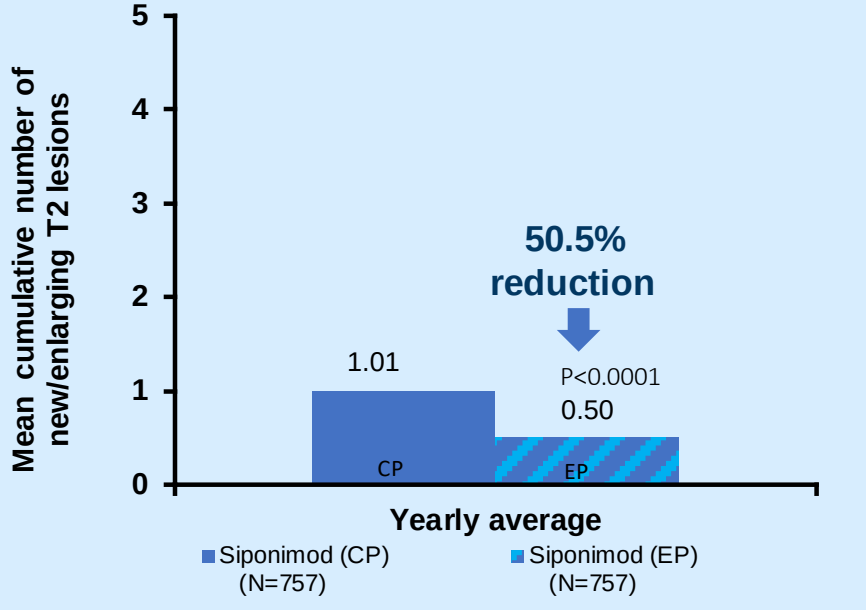
Mean cumulative number of new/enlarging T2 lesions
5
4
3
2
1
0
50.5% reduction
1.01
P<0.0001
0.50
CP
EP
Yearly average
Siponimod (CP) (N=757)
Siponimod (EP) (N=757)

- In the continuous siponimod group, benefit on all MRI measures was maintained during the extension part
- The low levels of BV loss, thalamic volume loss and T2 lesion activity observed in the core part were significantly lowered further in the extension part

At each time point, only patients with a value both at the CP visit and the corresponding EP visit are included; the yearly average is derived by standardizing the change to the last visit to 360 days
p-values from Wilcoxon signed-rank test comparing percent changes between CP and EP within each group

ARBA, annualized rate of brain atrophy; BV, brain volume; cGM, cortical gray matter; CP, core part; EP, extension part; MRI, magnetic resonance imaging; N, number of patients

Arnold et al.

Conclusions

- Siponimod treatment showed sustained efficacy on MRI parameters of inflammation and neurodegeneration (brain tissue atrophy including GM) over the long term in the EXPAND SPMS population
- In the continuous siponimod group, the low rate of brain tissue atrophy and lesion activity were maintained, and some parameters were significantly reduced further during the extension part, suggesting continued efficacy on those MRI outcomes over the long term
- Patients switching from placebo to siponimod in the extension part recapitulated pronounced reductions in the rate of brain atrophy and lesion activity with a complete suppression of cGM atrophy and no increase in T2 lesion volume within 12 months of starting siponimod
- Persistent differences between continuous and switch groups in measures of brain tissue integrity highlight the importance of early treatment initiation

Disclosures

DLA has received compensation from Alexion, Biogen, Celgene, Frequency Therapeutics, GENeuro, Genentech, Merck, Novartis, Receptos/Celgene, Roche, Sanofi; has received stock or an ownership interest from NeuroRx. His institution has received research support from Novartis and Immunotec. LK's institution has received research support from Bayer, Biogen, Celgene, Genentech, Gemzyme, Janssen, Merck Serono, Minoryx, Novartis, Roche, Sanolfi, Santhera, TG Therapeutics, Abbvie, Eisai, Swiss MS Society, Swiss National Research Foundation, European Union, Roche Research Foundation, Innosuisse. PV has received compensation from Biogen, Merck, Sanofi, Novartis, Roche, Sanofi, Teva, Celgene, Imcyse, AB Science and La Revue des Microbiotes. HIs institution has received research support from Roche and Sanofi. RG has received compensation from Biogen, Novartis, Genzyme, Bayer Vital, Eisai Pharmaceuticals, Roche, SAGE Publishers. His institution has received research support from Novartis and Biogen. AB-O has received compensation from Roche Genentech, Novartis, Janssen/Actelion, Atara Biotherapeutics, Biogen, BMS, Merk/EMD Serono, Sanofi-Genzyme. HI institution has received research support from Novartis, Biogen, Roche/Genentech, Merck/EMD Serono. GG has received compensation from Abbvie, Atara Bio, Biogen, Sanofi-Genzyme, Merck KGaA, Novartis, Roche, Actelion, Celgene, Roche, Medscape, Oxford Health Policy Forum,Neurology Academy, Peervoice, Elsevier and Bristows. BACC's institution has received compensation from Atara, Akili, Alexion, Biogen, EMD Serono, Novartis, Sanofi, TG Therapeutics. He has received publishing royalties from a publication relating to health care. RJF has received personal compensation from AB Science, Janssen, Biogen, Celgene, EMD Serono, Genentech, Immunic, Novartis, Sanofi, Teva, TG Therapeutics. His institution has received research support from National Institutes of Health. He has received publishing royalties from a publication relating to health care. DPM, SR, GK, FD and TH are employees of Novartis and may hold stocks of Novartis.

This study is funded by Novartis Pharma AG.

Acknowledgments: Writing support was provided by Shyamashree Dasgupta and Paul Coyle (Employees of Novartis). The final responsibility for the content lies with the authors.

Affiliations

[1]Brain Imaging Centre, Montreal Neurological Institute and Hospital, McGill University, Montreal, QC, Canada; [2]NeuroRx Research, Montreal, Quebec, Canada; [3]Neurologic Clinic and Policlinic and Research Center for Clinical Neuroimmunology and Neuroscience, Departments of Medicine, Clinical Research, Biomedicine and Biomedical Engineering, University Hospital and University of Basel, Basel, Switzerland; [4]Univ. Lille, Inserm UMR U1172 LilNCog, CHU Lille, FHU Precise, Lille, France; [5]Department of Neurology, St Josef−Hospital/Ruhr−University Bochum, Bochum, Germany; [6]Center for Neuroinflammation and Experimental Therapeutics and Department of Neurology, Perelman School of Medicine, University of Pennsylvania, Philadelphia, PA, USA; [7]Blizard Institute, Barts and The London School of Medicine and Dentistry, Queen Mary University of London, London, UK; [8]UCSF Weill Institute for Neurosciences, Department of Neurology, University of California San Francisco, San Francisco, CA, USA; [9]Novartis Pharma AG, Basel, Switzerland; [10]Novartis Pharmaceutical Corporation, East Hanover, NJ, USA; [11]Mellen Center for Treatment and Research in Multiple Sclerosis, Neurological Institute, Cleveland, OH, USA

Poster presented at the American Academy of Neurology (AAN) 2021, April 17-22, 2021